Predictors of response to a modified whole tumor cell immunotherapy in patients with advanced breast cancer from two phase I/IIa trials

William V. Williams[1], Markus D. Lacher[1], Daniel L. Adams[2], Shaker R. Dakhil[3], Jarrod Holmes[4], Saveri Bhattacharya[5], Carmen J. Calfa[6], George E. Peoples[7], and Charles L. Wiseman[1].

[1]BriaCell Therapeutics Corporation, Berkeley, CA; [2]Creatv MicroTech, Rockville, MD; [3]Cancer Center of Kansas, KS; [4]St. Joseph Health Santa Rosa, Santa Rosa, CA; [5]Thomas Jefferson University, Philadelphia, PA; [6]University of Miami Health, Miami, FL; [7]Cancer Insight LLC, San Antonio, TX

BriaCell

AACR 2020
Abstract CT102

ABSTRACT

Objectives: SV-BR-1-GM is a GM-CSF secreting breast cancer cell line derived from a Grade II (moderately differentiated) breast tumor that also expresses HLA class I & II antigens and is able to function as an antigen-presenting cell. Irradiated SV-BR-1-GM is used in a regimen including pre-dose low-dose cyclophosphamide and post-dose local IFN-α2b. The SV-BR-1-GM regimen has been used alone (“monotherapy”, ClinicalTrials.gov NCT03066947 – study completed) and in combination with checkpoint inhibitors (“combination”, ClinicalTrials.gov NCT03328026 – study ongoing). The objective is to evaluate potential predictors of response in advanced metastatic breast cancer (aMBC).

Methodology: 23 patients with refractory aMBC were treated with the SV-BR-1-GM regimen as monotherapy (cycles every 2 weeks x3 and then monthly). The combination study uses the SV-BR-1-GM regimen with PD-1 inhibitors pembrolizumab or INCMGA00012 with cycles every 3 weeks (12 patients dosed to date). Analyses include circulating tumor cells (CTCs), cancer-associated macrophage-like cells (CAMLs), tumor Grade (well- (Grade I), moderately- (Grade II) or poorly differentiated (Grade III)), HLA-type, and the ability to develop delayed-type hypersensitivity (DTH).

Preliminary Data: The patients were heavily pre-treated (median of 4 prior systemic therapies not including hormonal therapy). In the monotherapy study, disease control (including SD, PR or CR) was seen in 6 patients (26%, all with SD). 16 patients (76%) developed measurable DTH (2 patients with no data). In the combination therapy study, disease control was seen in 3 patients (25%, 1 PR and 2 SD) and 10 (91%) developed DTH. In the combined studies, patients with Grade I or Grade II tumors were more likely to achieve disease control (6/9, 67%) compared with those with Grade III tumors (2/20, 10%). Median progression free survival (PFS) on monotherapy was 2.6 months and 4.5 months for patients with Grade I/II disease. Median PFS on the combination study was 4.2 months and 6.9 months for patients with Grade I/II tumors. Patients with 1 or 2 HLA matches with SV-BR-1-GM achieved disease control in 29% (6/21) and 38% (5/13), respectively. In patients with Grade I or II tumors, for those with 1+ or 2+ HLA matches, disease control was seen in 57% (4/7) and 75% (3/4), respectively. None of the 3 patients with high levels of CTCs at baseline had disease control. Mean and median PD-L1 expression levels on CTCs and CAMLS were higher for patients with Grade III tumors compared with Grade I/II tumors, but this was not clearly correlated with clinical response.

The SV-BR-1-GM regimen can produce disease control and clinical benefit in very heavily pretreated patients with aMBC refractory to conventional therapies especially if the patients have an intact immune system (DTH), do not have high levels of CTCs, have Grade I or II disease, and have HLA matching with the SV-BR-1-GM cell line.

BACKGROUND

- SV-BR-1-GM is a whole-cell, GM-CSF expressing breast cancer cell line (**derived from a grade II tumor**) with features of antigen-presenting cells including HLA class II expression.
- In an **initial, pilot Phase I** clinical trial with 4 evaluable subjects, one “Special Responder” experienced prompt, widespread regression at multiple sites of metastatic breast cancer (Wiseman and Kharazi, 2006; The Breast Journal, Volume 12 Number 5, 2006 475– 480).
- In a **completed Phase I/IIa** clinical trial for advanced breast cancer (ClinicalTrials.gov NCT03066947) with 23 subjects dosed with SV-BR-1-GM, tumor regression was noted in several subjects. The clinical benefit rate for Grade I/II patients with immune responses (DTH) was 5/7 (71%).
- In an **ongoing Phase I/IIa** clinical trial for advanced breast cancer testing SV-BR-1-GM **in combination with a PD-1 inhibitor** (pembrolizumab/Keytruda, Merck; or INCMGA00012, Incyte/Macrogenics) (ClinicalTrials.gov NCT03328026), 12 subjects have thus far been dosed. Three of four patients with Grade I/II tumors had disease control (75%).

Mechanism of Action (MoA)
SV-BR-1-GM acts as an antigen-presenting cell for primed T cells (Lacher *et al.*, Front Immunol. 2018 May 15;9:776 and **Figure 1**).

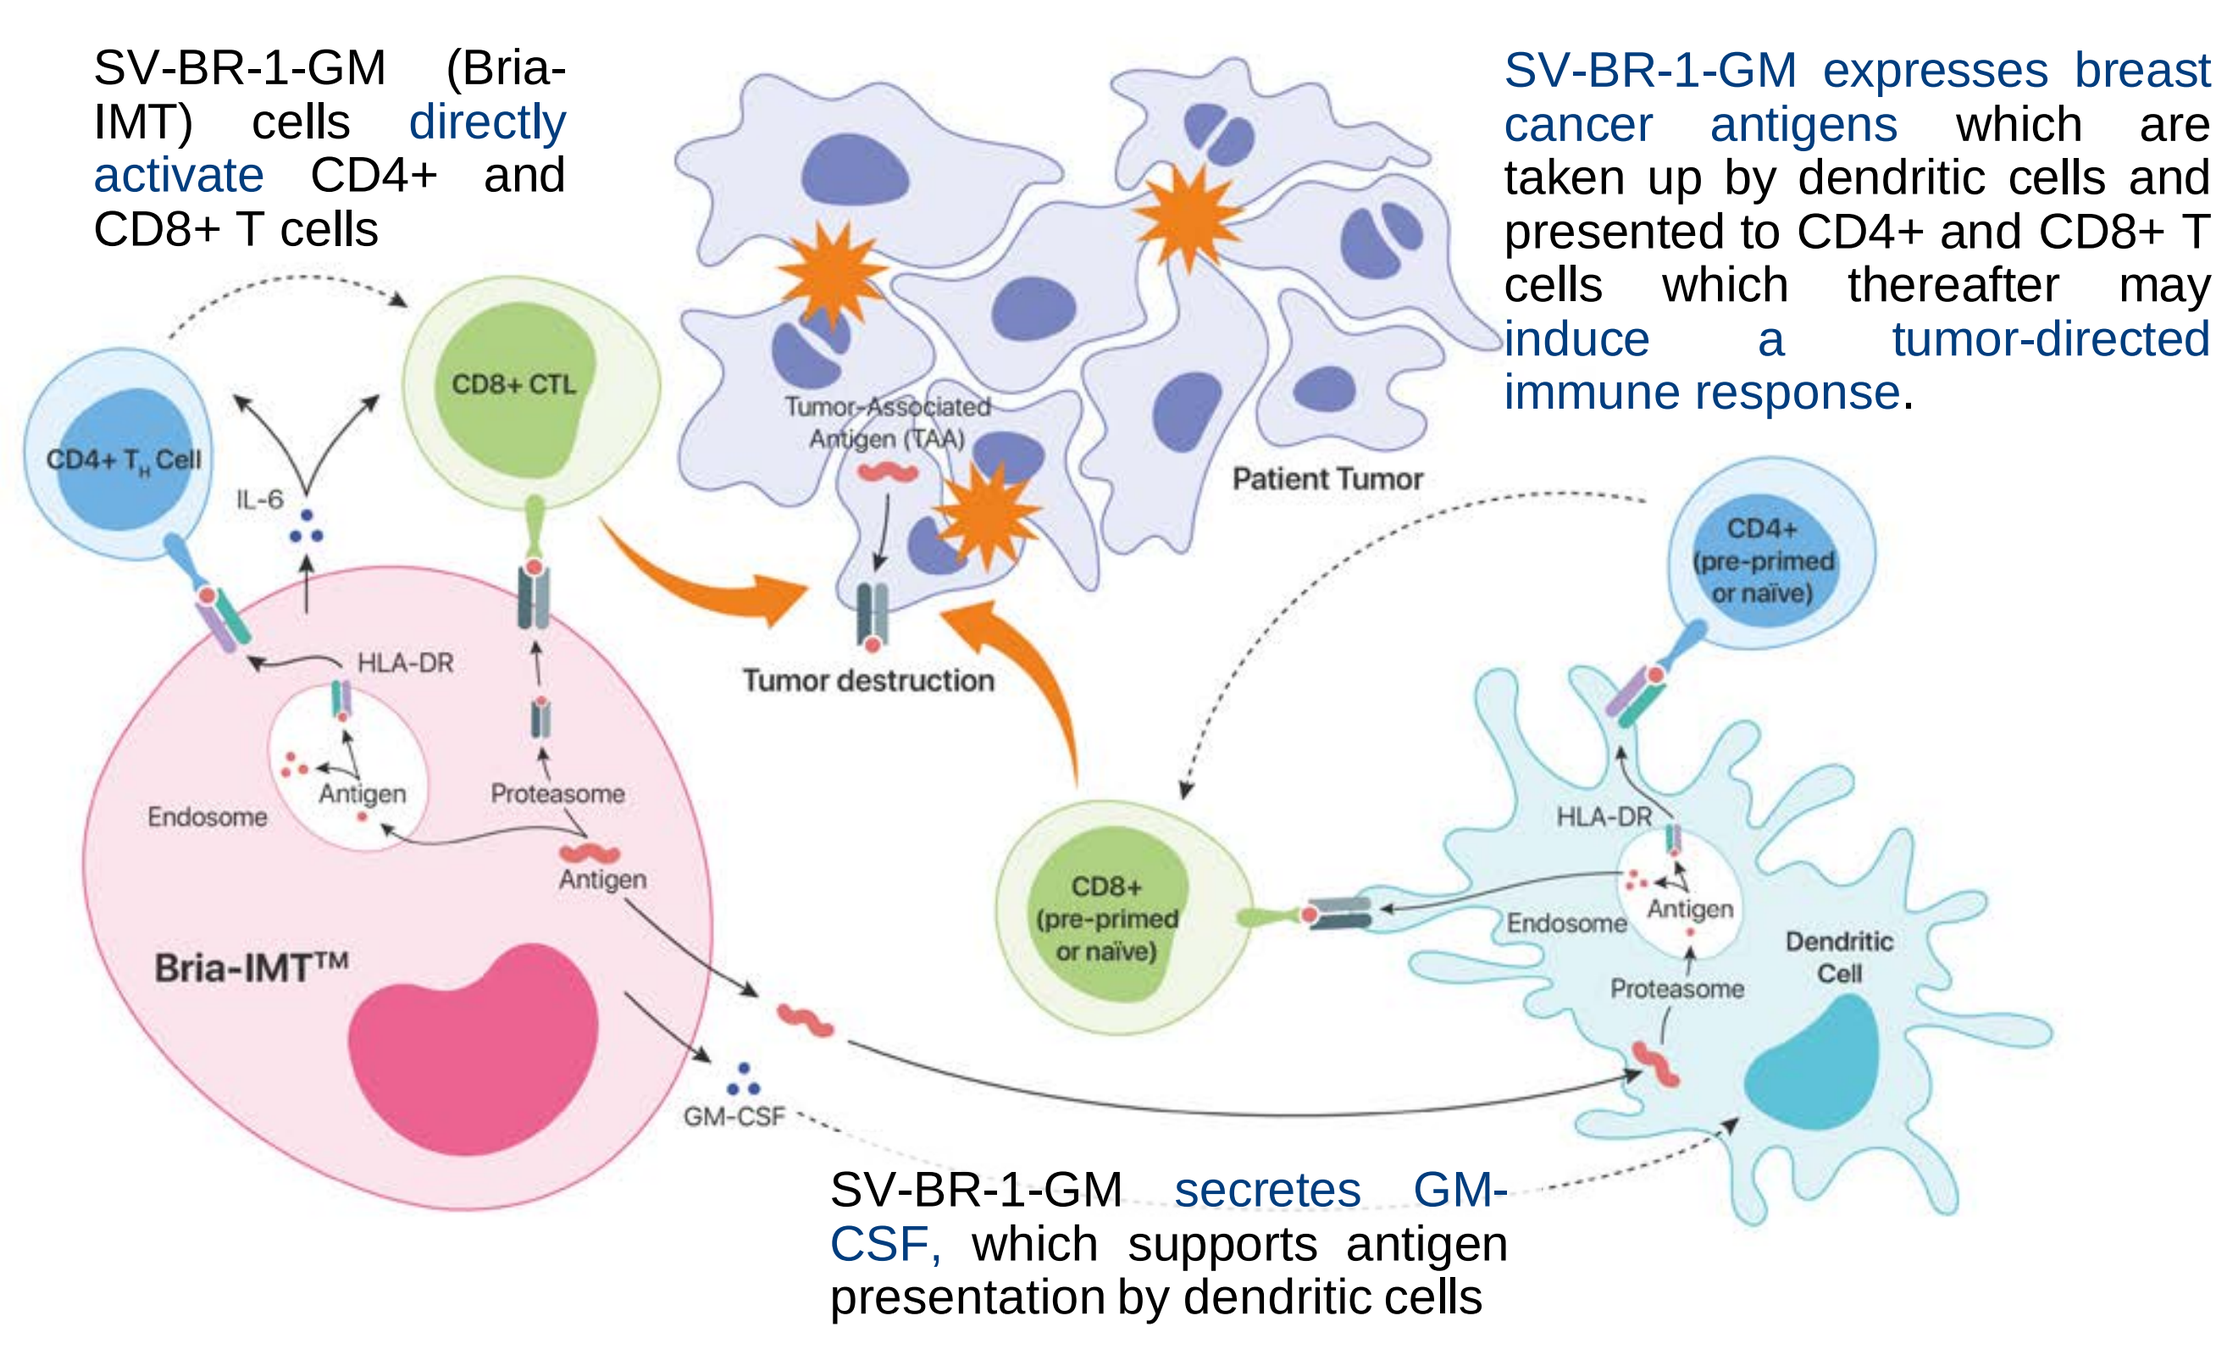
SV-BR-1-GM (Bria-IMT) cells directly activate CD4+ and CD8+ T cells
SV-BR-1-GM expresses breast cancer antigens which are taken up by dendritic cells and presented to CD4+ and CD8+ T cells which thereafter may induce a tumor-directed immune response.
SV-BR-1-GM secretes GM-CSF, which supports antigen presentation by dendritic cells
CD4+ T_H Cell
CD8+ CTL
IL-6
HLA-DR
Endosome
Antigen
Proteasome
Bria-IMT™
GM-CSF
Tumor-Associated Antigen (TAA)
Patient Tumor
Tumor destruction
CD8+ (pre-primed or naïve)
CD4+ (pre-primed or naïve)
Dendritic Cell

Figure 1. Model of Proposed MoA of SV-BR-1-GM

METHODS

Patient Treatment

The SV-BR-1-GM “Monotherapy” Regimen:

- Pre-dose cyclophosphamide (300 mg/m^2) 2-3 days prior to SV-BR-1-GM inoculation.
- DTH skin test with ~1 million cells (forearm), then inoculation of **~20**-50 million irradiated SV-BR-1-GM cells intradermally, in 4 inoculations (x2 upper back, x2 thighs).
- Interferon-α2b intradermally (10,000 IU in each inoculation site) 2±1 and 4±1 days following SV-BR-1-GM inoculation.
- PD-1i Combination Therapy only: Pembrolizumab (Keytruda; 200 mg IV) or INCMGA00012 (375 mg IV) during one of the IFN-a2b visits.

Cycles:

- NCT03066947 (“monotherapy”): Treatment is performed every 2 weeks for the first month and then every month.
- NCT03328026 (PD-1i combination with pembrolizumab or INCMGA00012): Treatment every 3 weeks.

RESULTS

	Patient Characteristics by Grade		
Study	Grade I/II (n=9)	Grade III (n=20)	All (n=31)
Monotherapy (n=23)	6	17	23
Combination Therapy (n=12)	4	7	12
Age	63 ± 8	56 ± 10	58 ± 10
Median Prior Systemic Regimens	8 (range 0-10)	4 (range 1-10)	5 (range 0-12)
Median Prior Hormonal Regimens	1 (range 0-5)	0 (range 0-5)	0 (range 0-5)
% ER/PR +	89%	48%	60%
% Her2/neu +	22%	5%	10%
% Triple Negative	11%	48%	37%
PD-L1 Expression on CTCs Mean ± SEM (Median)	211 ± 118 (122)	582 ± 275 (330)	417 ± 166 (319)
Disease Control (SD, PR or CR)	6/9 (67%)	2/20 (10%)	8/31 (26%)
Disease Control in DTH Responders	6/8 (75%)	2/14 (14%)	8/22 (36%)
Disease Control: No HLA Matches	2/2 (100%)	0/6 (0%)	2/8 (25%)
Disease Control: 1+ HLA Match	4/6 (67%)	2/13 (15%)	8/26 (31%)
Disease Control: 2+ HLA Matches	4/5 (80%)	1/6 (17%)	5/11 (45%)
Circulating Tumor Cells (CTCs) >100	-	0/3 (0%)	0/3 (0%)

Table 1. Patient Characteristics from the “Monotherapy” Study and the PD-1 Inhibitor Combination Study by Tumor Grade. The patients were all heavily pretreated having failed multiple prior regimens.

CONCLUSIONS AND HYPOTHESES

- **The SV-BR-1-GM regimen +/- PD-1i is able to induce an effective immune response and disease control in heavily pre-treated advanced breast cancer patients.**
- **Patients with grade I (well-differentiated) or grade II (moderately differentiated) tumors are more likely to respond with disease control and longer progression free survival.**
- **A more robust immune response, as noted by DTH, appears to correlate with disease control and longer progression free survival.**
- **Matching the SV-BR-1-GM cell line by 2+ HLA alleles appears to correlate with enhanced disease control and PFS in the monotherapy study, but not in the PD-1i combination study.**
- **None of 3 patients with elevated circulating tumor cells displayed disease control.**
- **Further study of the SV-BR-1 regimen is warranted especially in these patient populations.**

RESULTS

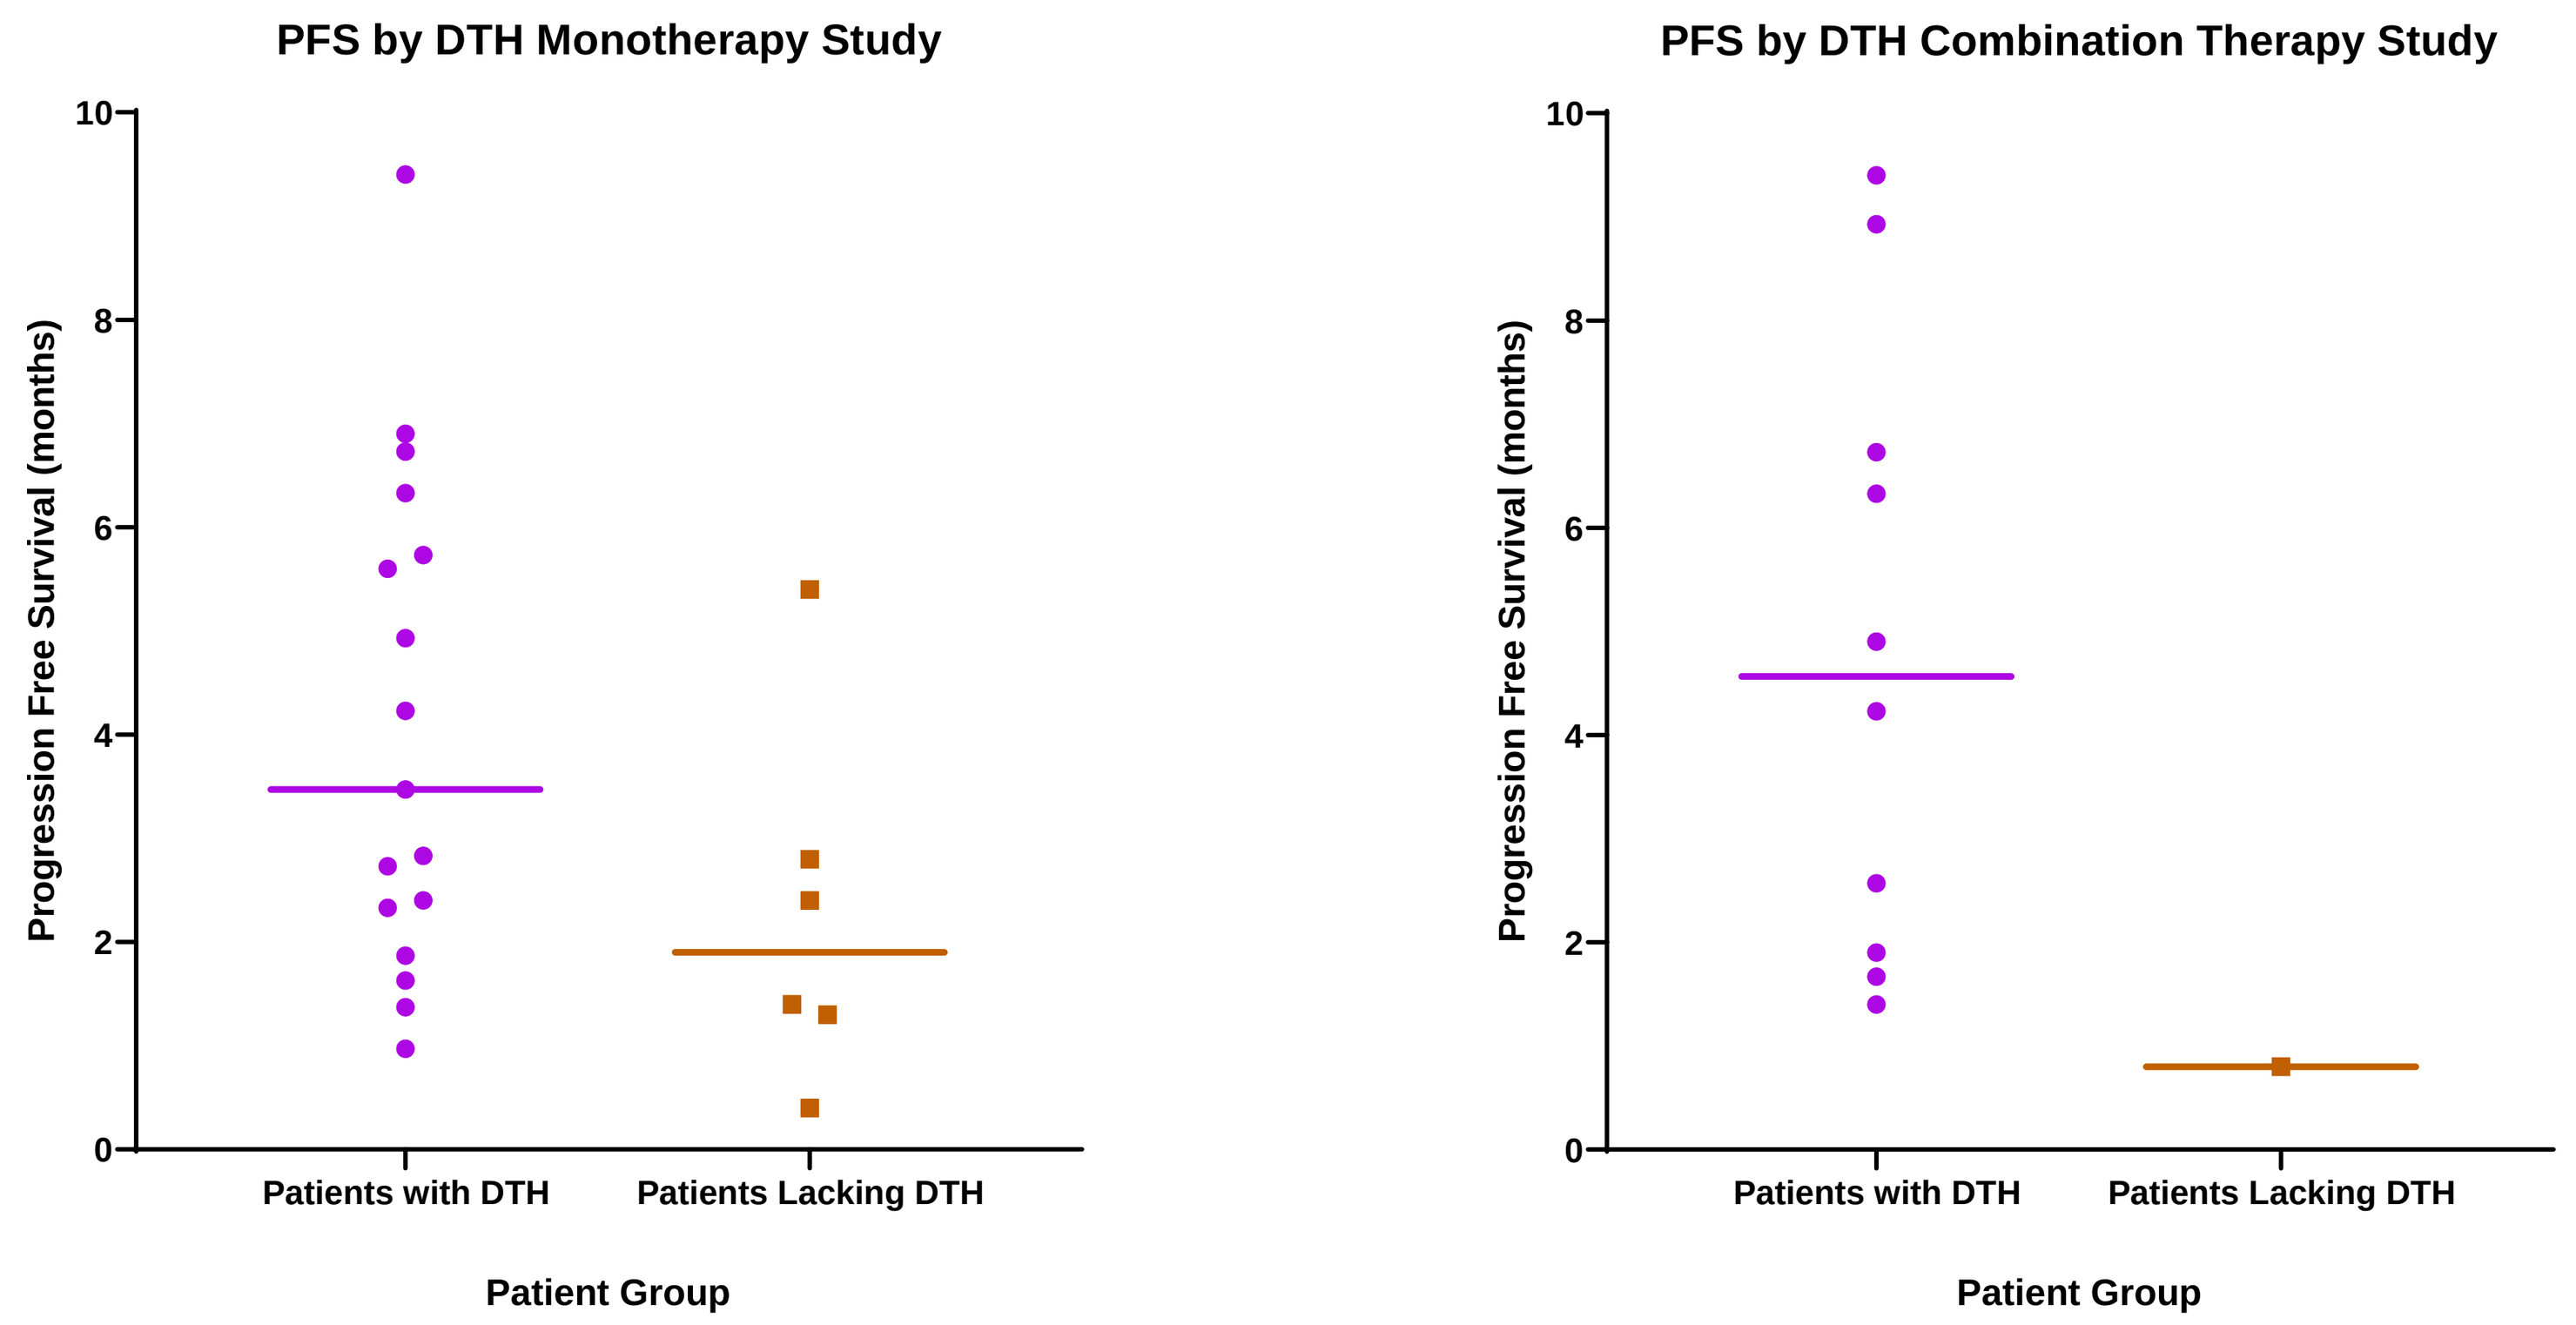
PFS by DTH Monotherapy Study
PFS by DTH Combination Therapy Study
Progression Free Survival (months)
Patients with DTH
Patients Lacking DTH
Patient Group

Figure 2. Delayed Type Hypersensitivity (DTH) and Progression Free Survival (PFS). For both the monotherapy study (left) and the PD-1i combination therapy study (right), patients who were able to develop measurable DTH to SV-BR-1-GM displayed higher disease control rates (Table 1) and longer progression-free survival. The changes in PFS were significant for all patients (both studies 2-tailed T.TEST, 2 sample with unequal variance) p <0.05.
Development of DTH appears to correlate with better response to therapy and PFS.

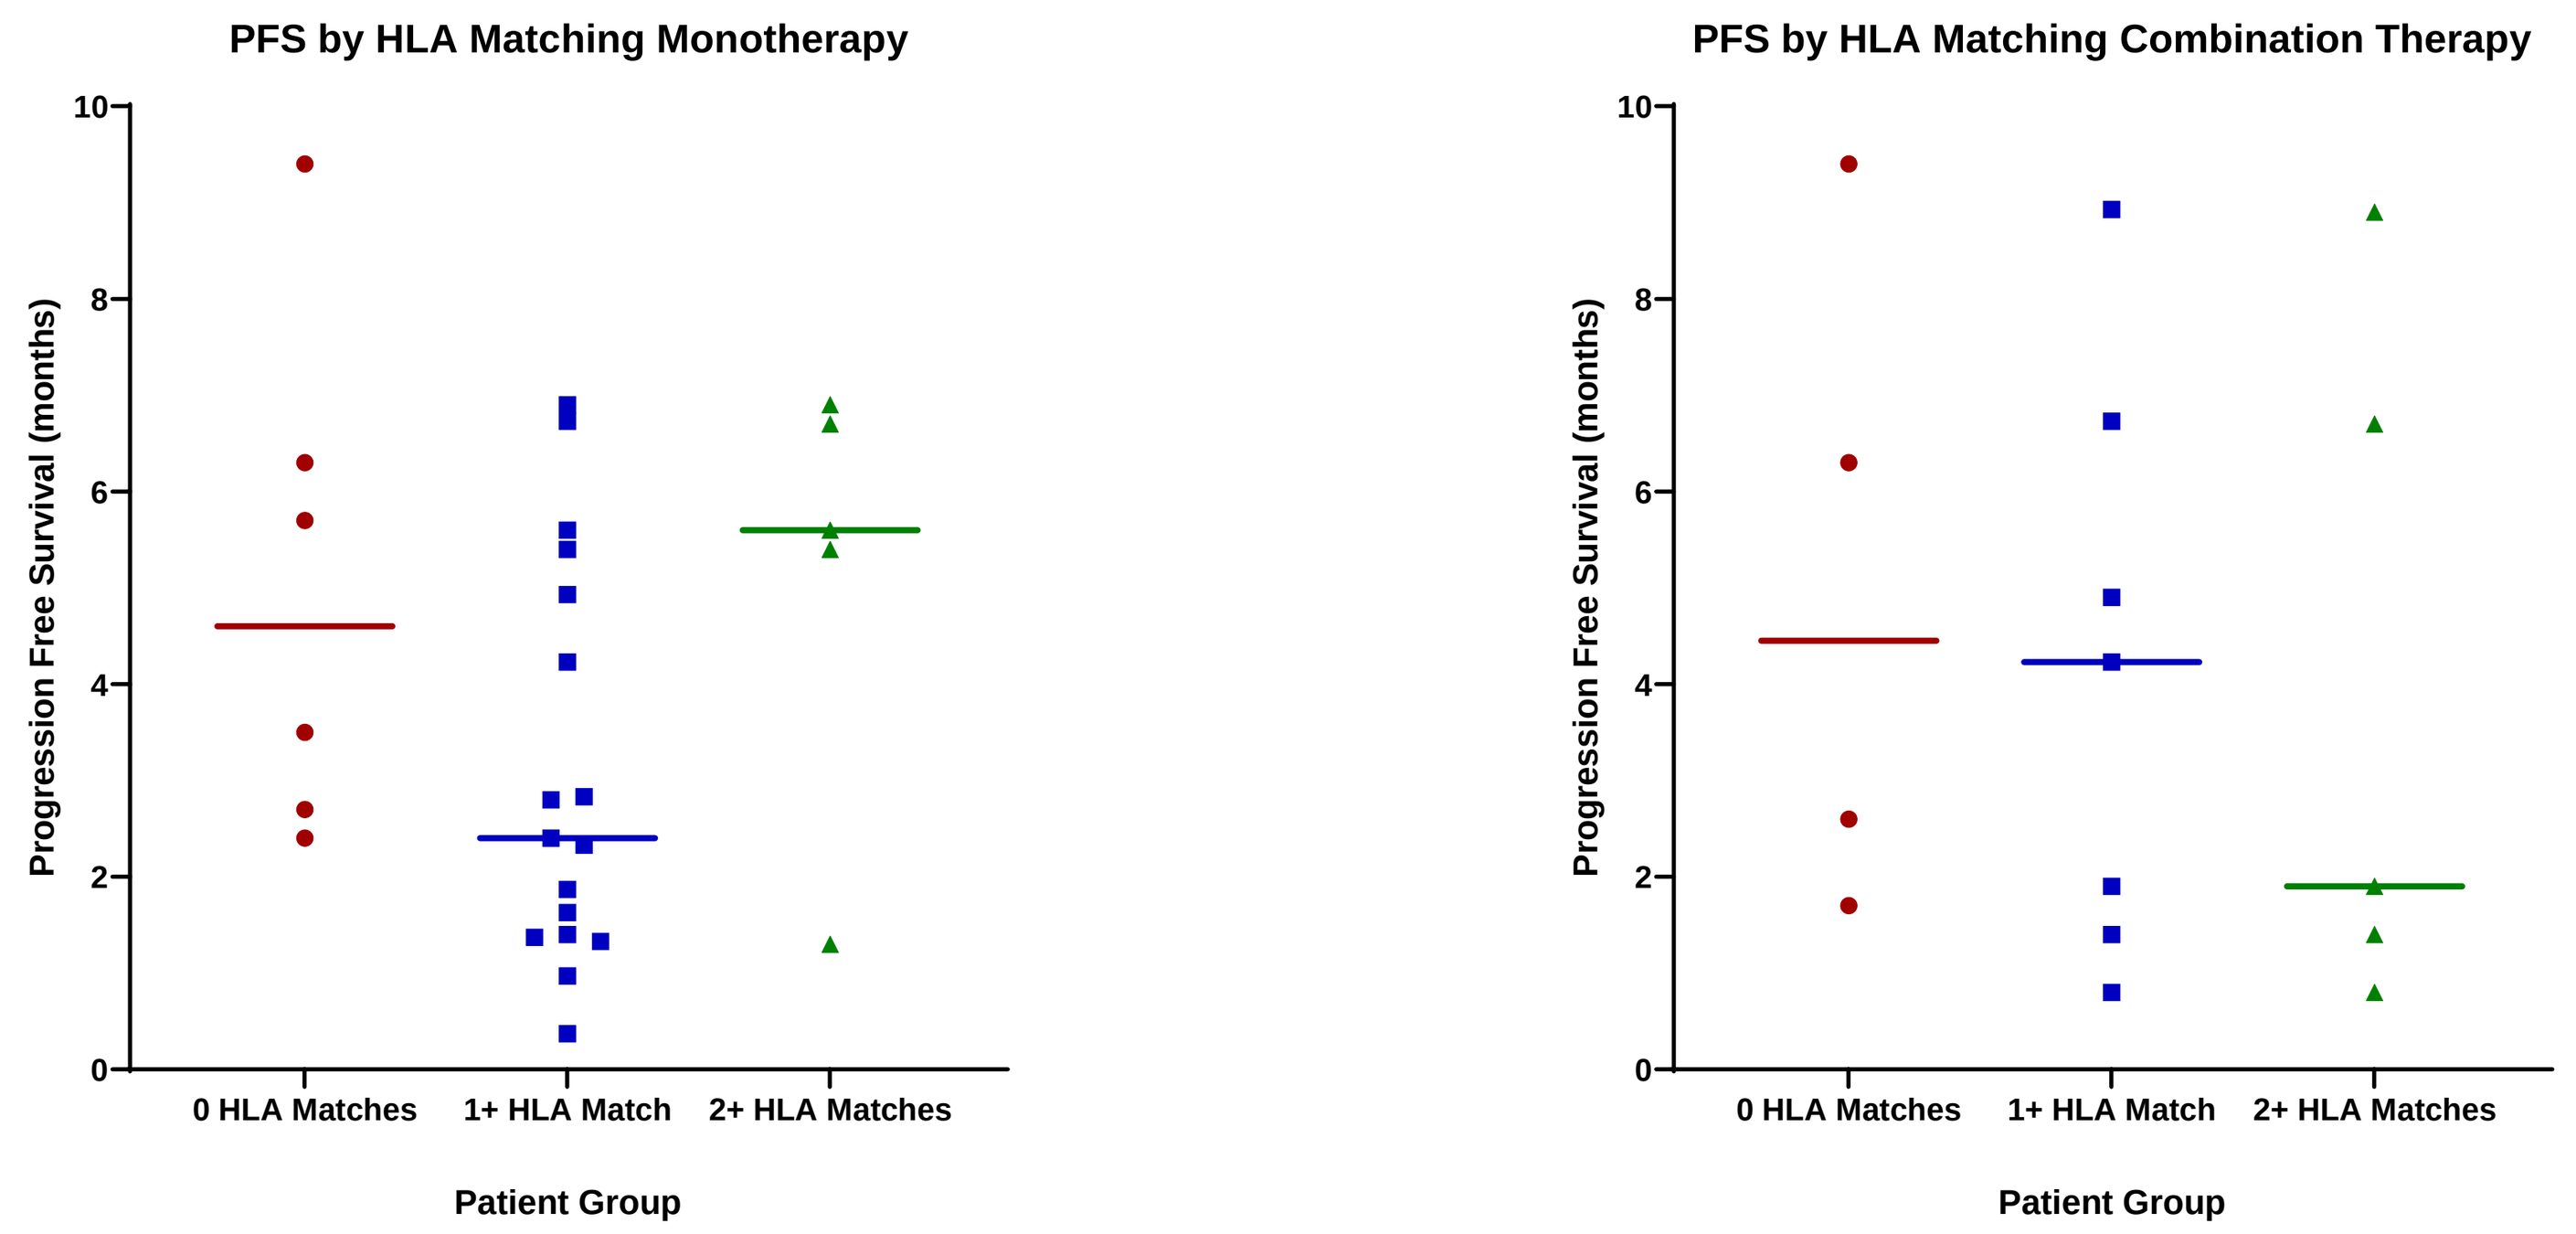
PFS by HLA Matching Monotherapy
PFS by HLA Matching Combination Therapy
Progression Free Survival (months)
0 HLA Matches
1+ HLA Match
2+ HLA Matches
Patient Group

Figure 3. HLA Matching with SV-BR-1-GM and Progression Free Survival (PFS). For the monotherapy study (left) patients with 2+ HLA matches with SV-BR-1-GM appear to have somewhat improved PFS and disease control rates (Table 1). But this is not the case for the PD-1i combination therapy study. No significant changes were noted. **HLA matching at 2+ loci appears to correlate with better response to therapy and PFS for monotherapy but not combination therapy.**

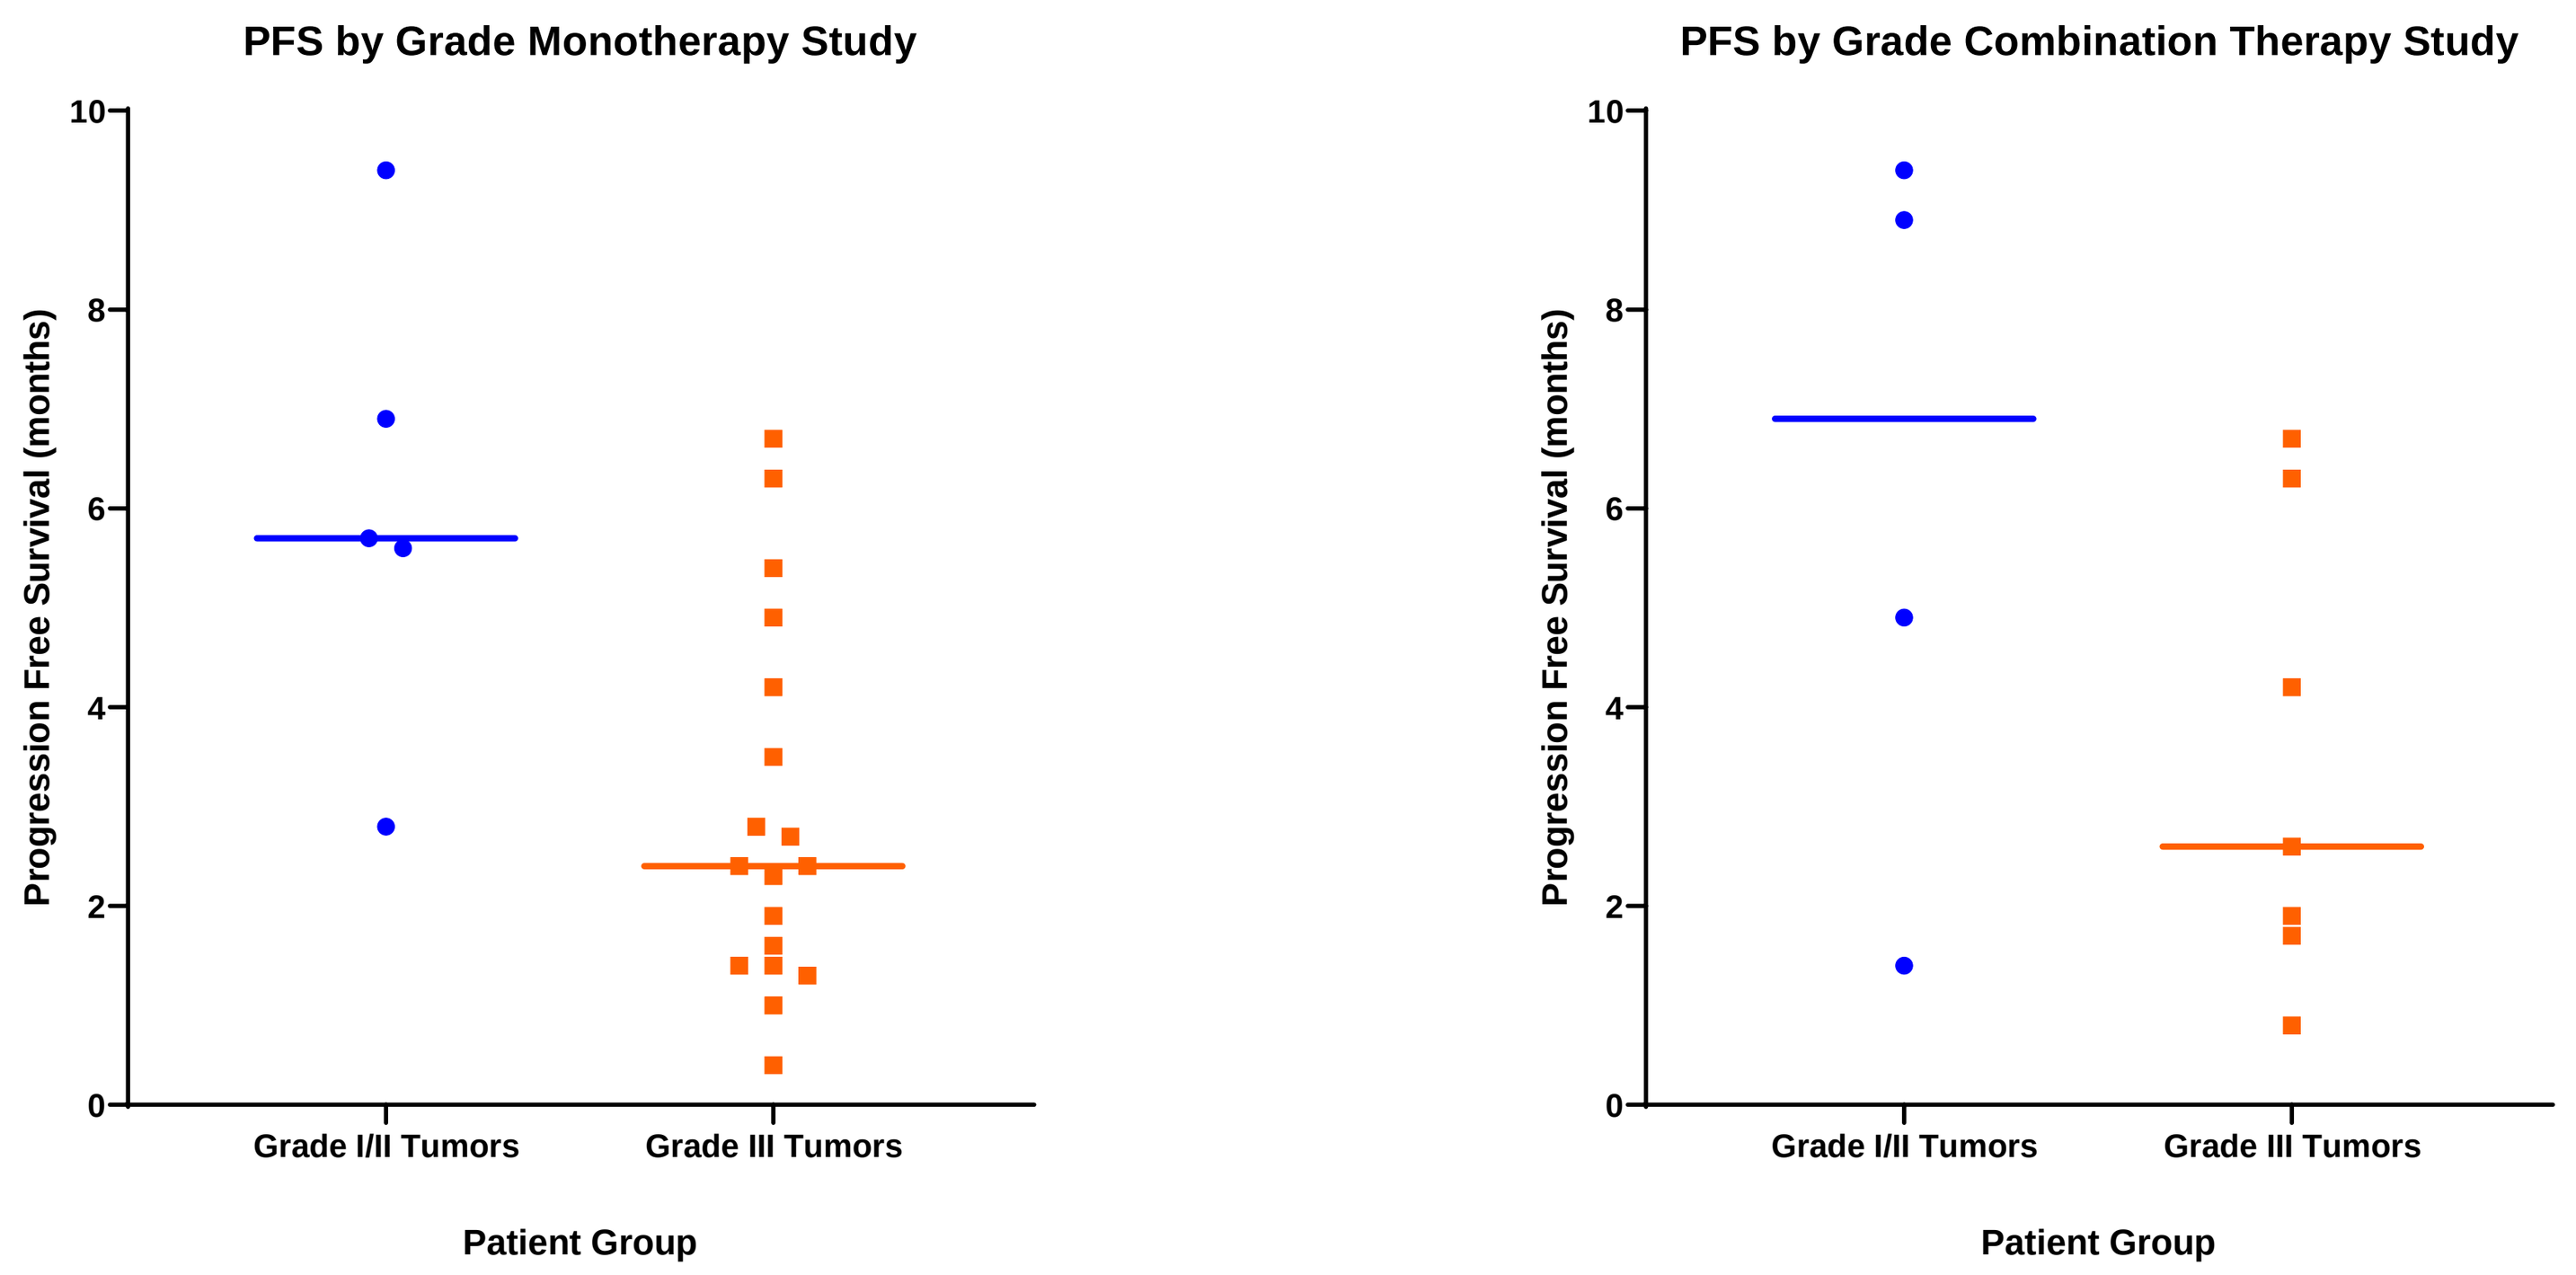
PFS by Grade Monotherapy Study
PFS by Grade Combination Therapy Study
Progression Free Survival (months)
Grade I/II Tumors
Grade III Tumors
Patient Group

Figure 4. Tumor Grade and Progression Free Survival (PFS). SV-BR-1-GM was derived initially from a biopsy of a grade II (moderately differentiated) metastatic breast cancer lesion. Therefore, we evaluated the PFS of patients with Grade I (well-differentiated, n=1) and Grade II (n=7) tumors compared with Grade III (poorly differentiated, n=22) tumors in the monotherapy study (left) and the PD-1i combination therapy study (right). The changes in PFS were significant for all patients (both studies) and for the monotherapy study (2-tailed T.TEST, 2 sample with unequal variance) p <0.05.
Grade I/II tumor histology appears to correlate with better response to therapy (Table 1) and PFS.

Copyright BriaCell Therapeutics Corp.

Template from: Call4Posters™